15th International Adenovirus Meeting

Abstract #16

circAde, a viral vector system for high and durable protein expression

O'Leary ET[1,2], Vikberg S[1,2], Foord E[1,2], Wiklund ED[1], Levitsky V[1], Hansen TB[1]

1. Circio AB, Stockholm, Sweden
2. Karolinska Institutet, CLINTEC, Huddinge, Sweden

Introduction

Circular RNAs (circRNAs) are naturally occurring transcripts found in all eukaryotes. CircRNAs typically derive from protein-coding exons prone to backsplicing (non-linear splicing in which an upstream splice acceptor attacks a downstream donor, see schematic below). In contrast to mRNAs, circRNAs are highly stable molecules with sustained effects within cells. Understanding the biogenesis and potential of circRNA allows repurposing the stable circRNA features into durable protein-coding templates. Here, we show our development of our vector-based system for efficient delivery and expression of circRNA.

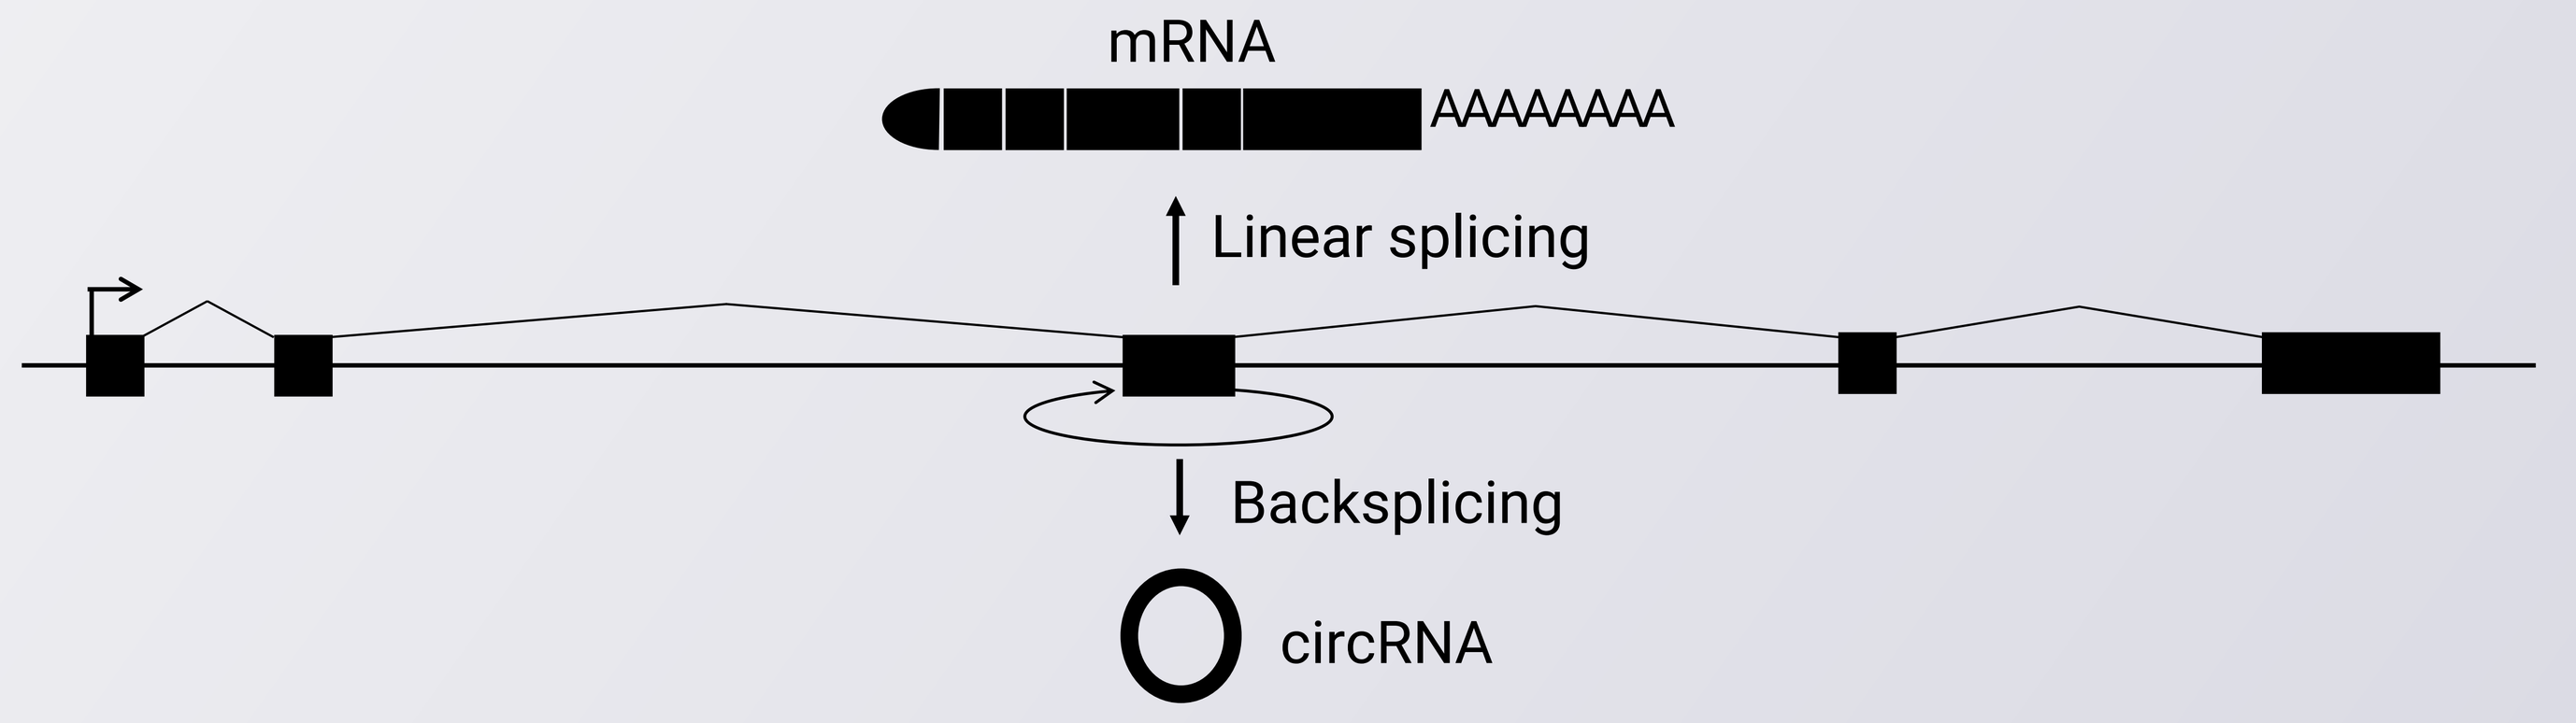
mRNA
AAAAAAAA
Linear splicing
Backsplicing
circRNA

Choice and composition of IR dramatically impacts circRNA biogenesis

Based on endogenous human loci with high-yield circRNA production, the prevalent model for circRNA biogenesis involves flanking inverted repeats (IRs). We show that the IRs are required for biogenesis (**Fig. 1A-C**), and that the choice of IR dramatically impacts circRNA yield. Here, bioinformatic analysis of publicly available RNA-Seq datasets was used to identify highly abundant circRNAs (**Fig. 1D)**. Several IR's flanking these abundant circRNAs were incorporated into our circRNA cassette to examine their ability to drive backsplicing (**Fig. 1E-G)**.

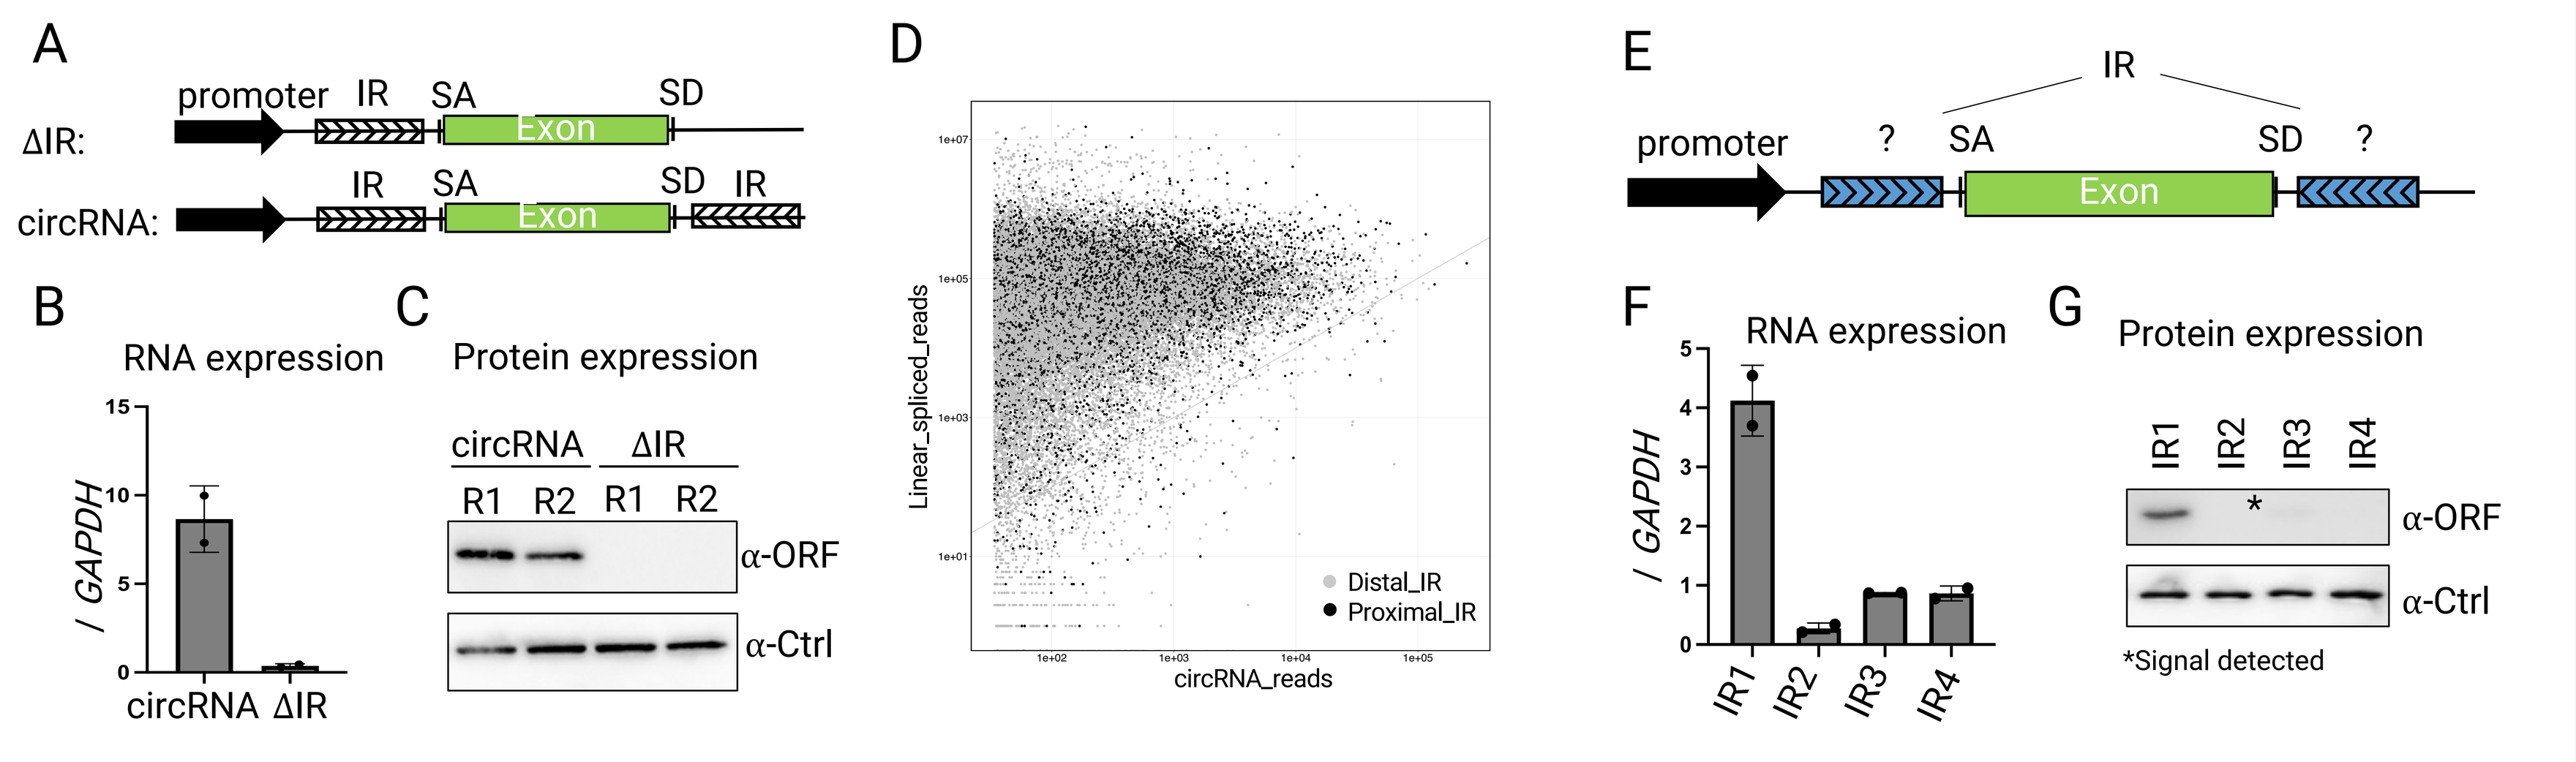
A
promoter IR SA SD
ΔIR:
Exon
circRNA:
IR SA SD IR
B
RNA expression
/ GAPDH
circRNA ΔIR
C
Protein expression
circRNA ΔIR
R1 R2 R1 R2
α-ORF
α-Ctrl
D
Linear_spliced_reads
circRNA_reads
Distal_IR
Proximal_IR
E
IR
promoter ? SA SD ?
Exon
F
RNA expression
/ GAPDH
IR1 IR2 IR3 IR4
G
Protein expression
IR1 IR2 IR3 IR4
α-ORF
α-Ctrl
*Signal detected

Figure 1: Optimization of flanking inverted repeats (IRs). A) Schematic representation of circRNA cassettes with or downstream inverted repeat (IR). **B-C)** Biogenesis as measured by qRT-PCR (B) and protein yield measured by western blotting (C) from circRNA cassettes with or without downstream IR. **D)** Schematic representation of circRNA cassettes with depicting IR position, where different proximal IR elements from highly expressed endogenous circRNAs are incorporated. **E-F)** Biogenesis as measured by qRT-PCR (E) and protein yield measured by western blotting (F) from circRNA cassettes with different IRs. **G)** The impact of downstream IR sequence composition on circRNA biogenesis as measured by qRT-PCR normalized to *GAPDH* mRNA.

Importance of cassette design for protein-coding circRNA

Endogenous circRNAs are not *per se* a template for translation. Thus, protein translation from circRNA requires an IRES (internal ribosomal entry site) for cap-independent translation (**Fig. 2A**). Here, testing 10 different designs using 2 different IRES elements (schematically shown in **Fig. 2B**), a dramatic design-dependent effect on biogenesis was observed by protein expression, with high and consistent yield from the circRNA-producing D4 and D5 designs (**Fig. 2C**).

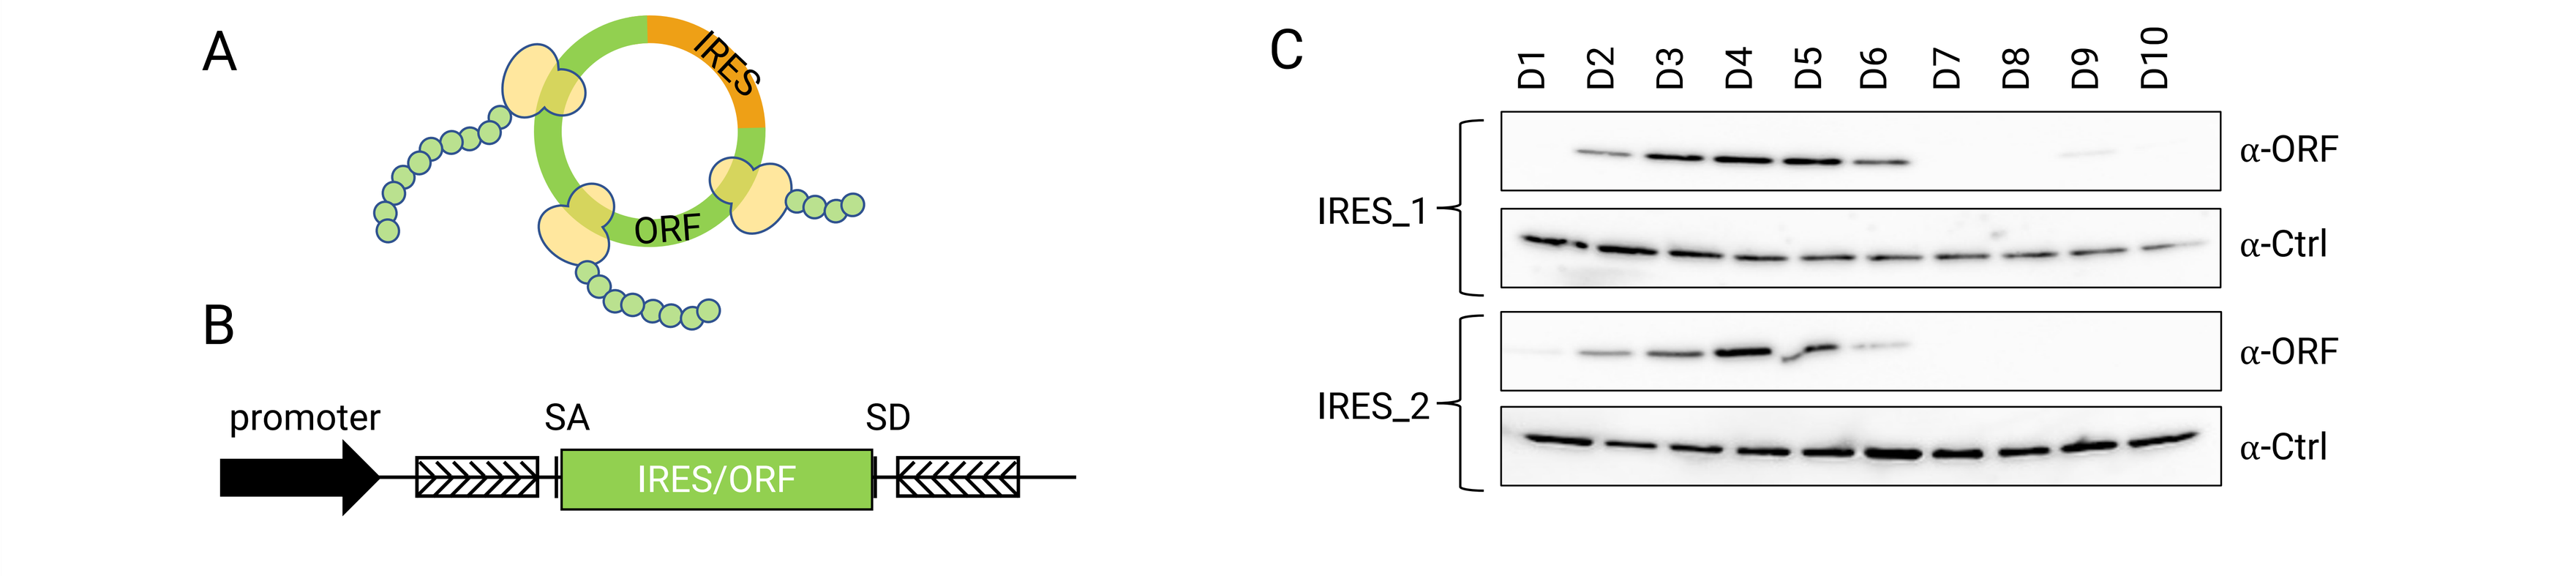
A
IRES
ORF
B
promoter SA SD
IRES/ORF
C
D1 D2 D3 D4 D5 D6 D7 D8 D9 D10
IRES_1
IRES_2
α-ORF
α-Ctrl

Figure 2: Choice of IRES and IRES/ORF composition impact circRNA expression: A) CircRNA translation depends on cap-independent translation and requires IRES (internal ribosome entry site) and ORF (Open Reading Frame) as shown. **B)** Schematic representation of vector cassette used for circRNA expression. **C)** Protein expression from ten different IRES/ORF designs (D1-D10) using two different IRES elements was assessed by western blot.

Enhanced circRNA stability confers prolonged protein expression

Almost all cellular RNA turnover is facilitated by exonucleolytic decay. Circular RNAs are devoid of 5' and 3' ends and thereby resistant to exonucleases. Consequently, high stability and long half-lives are observed for circRNA compared to mRNA (**Fig. 3A-B**). To benchmark the circRNA expression cassette with conventional mRNA-based expression vectors, the D4 circRNA comprising either IRES1 or IRES2 was compared to mRNA expression from the same backbone. The D4_IRES1 design showed superior protein yield 48 hours after transfection (**Fig. 3C-E**), despite reduced levels of RNA (**Fig. 3E**), suggesting that cap-independent IRES-mediated translation is more effective than cap-dependent translation.

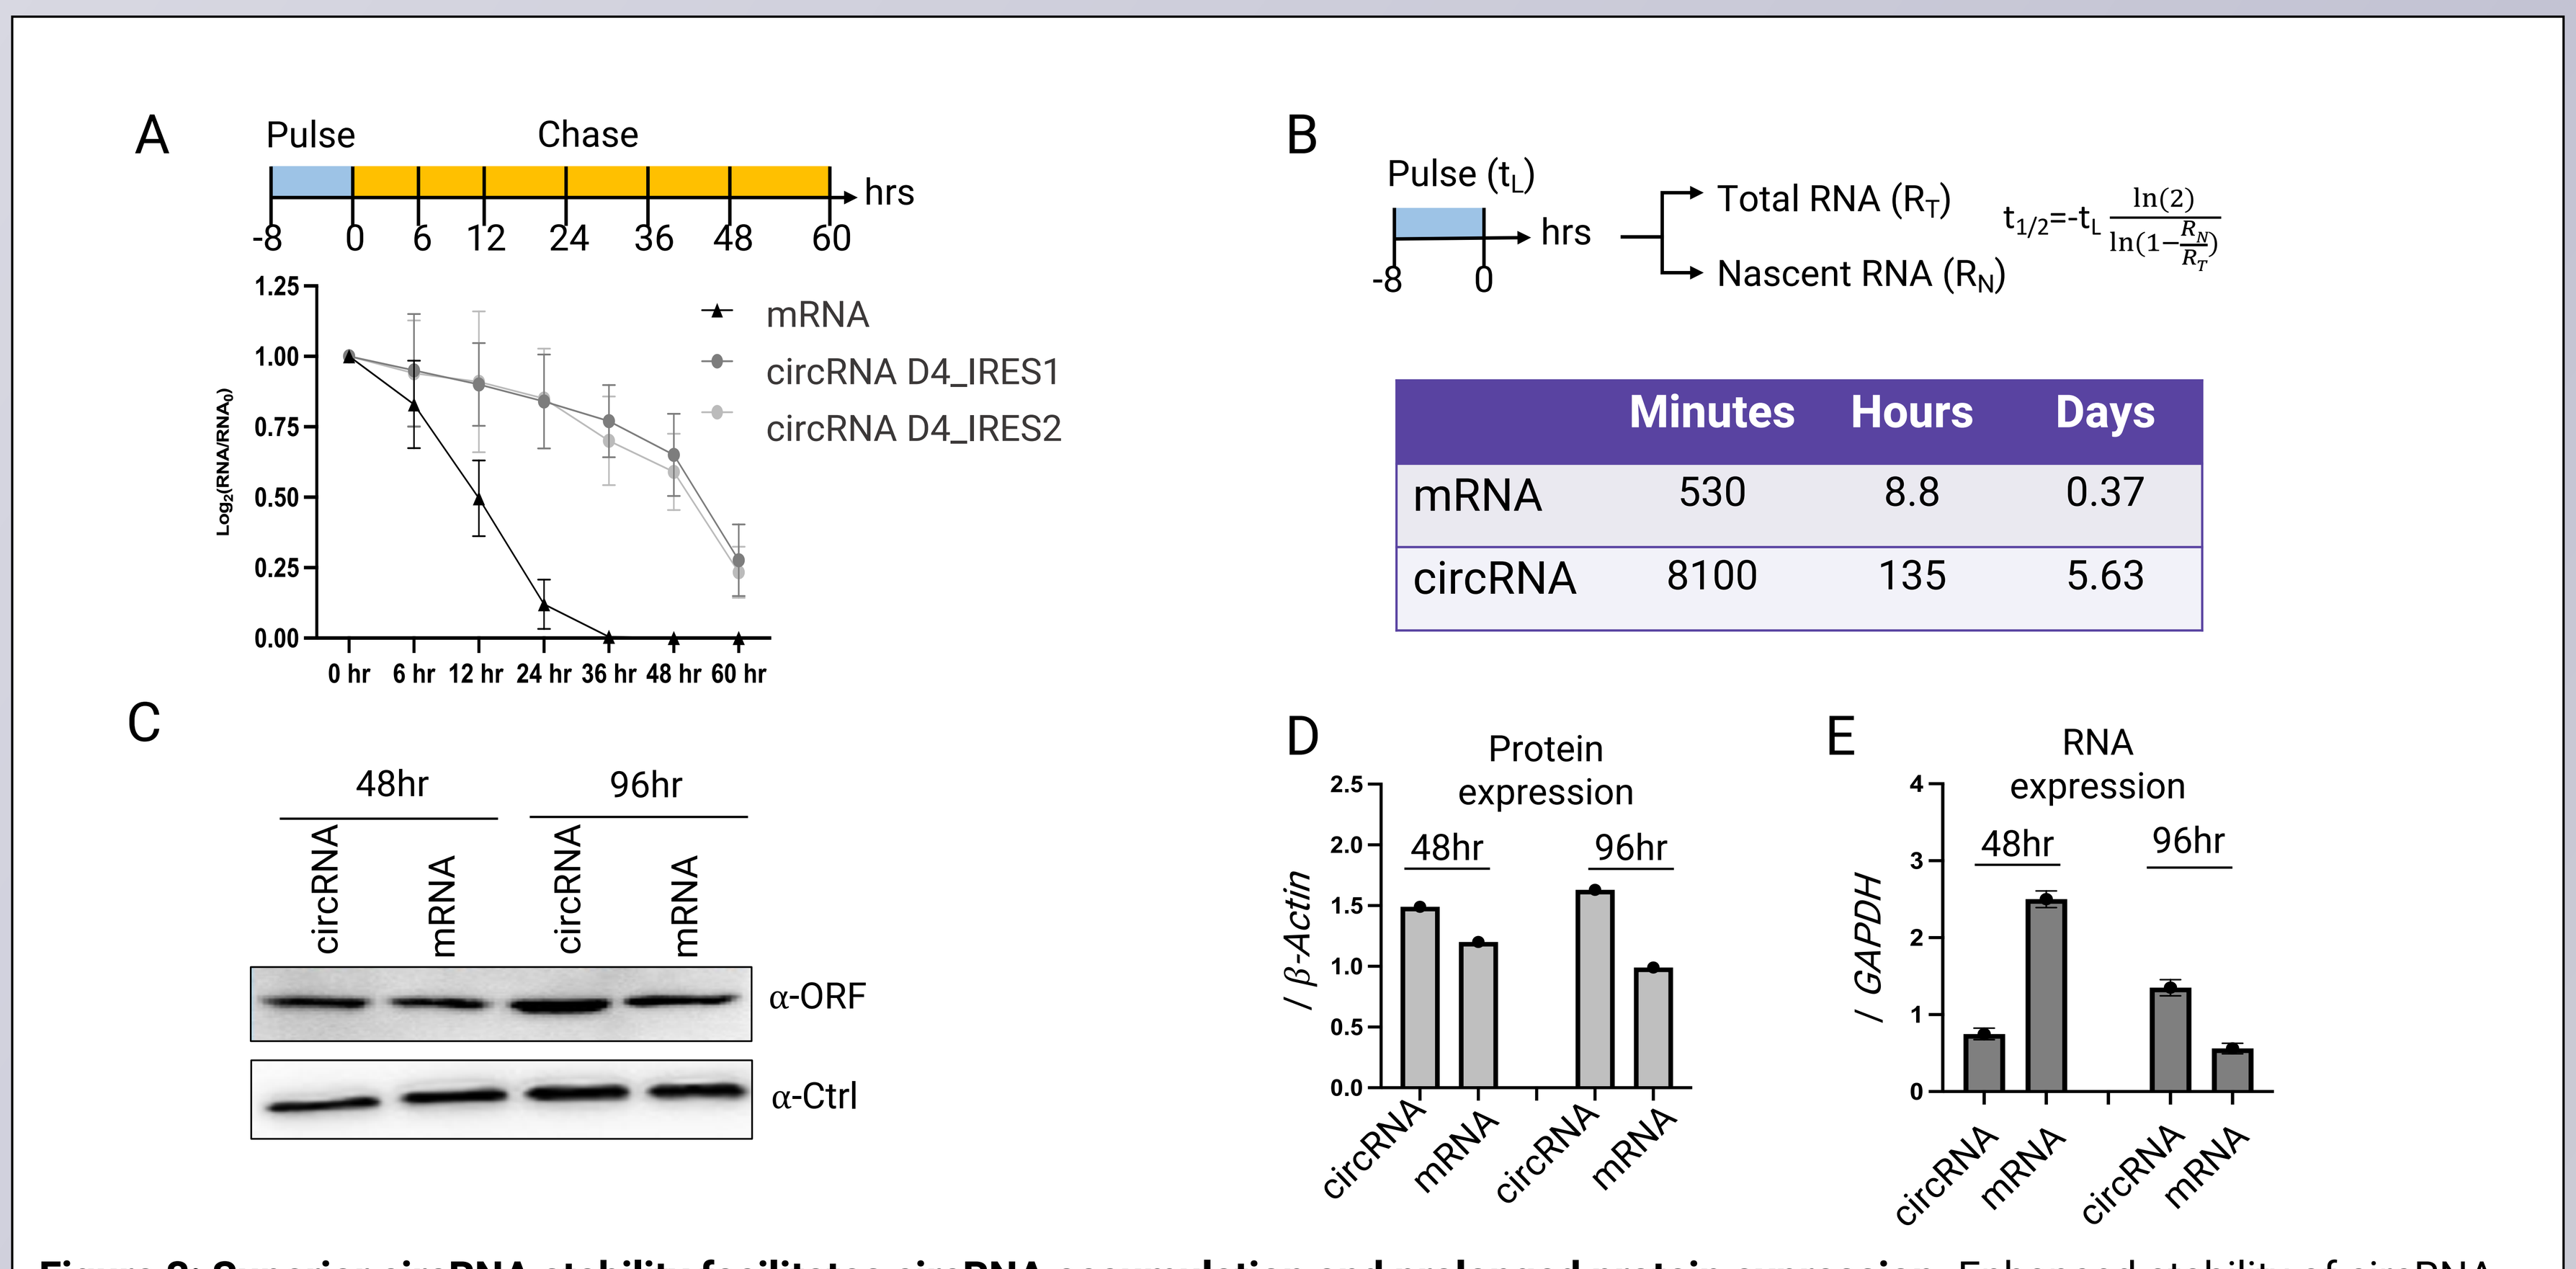
A
Pulse
Chase
hrs
-8 0 6 12 24 36 48 60
mRNA
circRNA D4_IRES1
circRNA D4_IRES2
0 hr 6 hr 12 hr 24 hr 36 hr 48 hr 60 hr
B
Pulse (t_L)
hrs
-8 0
Total RNA (R_T)
Nascent RNA (R_N)
$t_{1/2}=-t_L \frac{\ln(2)}{\ln(1-\frac{R_N}{R_T})}$
C
48hr 96hr
circRNA mRNA circRNA mRNA
α-ORF
α-Ctrl
D
Protein expression
/ β-Actin
48hr 96hr
circRNA mRNA circRNA mRNA
E
RNA expression
/ GAPDH
48hr 96hr
circRNA mRNA circRNA mRNA

	Minutes	Hours	Days
mRNA	530	8.8	0.37
circRNA	8100	135	5.63

Figure 3: Superior circRNA stability facilitates circRNA accumulation and prolonged protein expression. Enhanced stability of circRNA compared to mRNA was demonstrated using a pulse-chase experiments (A) and half-life estimations (B). **A)** Newly synthesized RNA was labelled with nucleotide analogues for 8 hours and the durability of labelled RNA was quantified over time by qRT-PCR. **B)** Half-life RNA estimates were inferred from the nascent fraction (newly synthesised labelled RNA as in(A)) relative to total RNA assuming steady-state**. C-E)** Quantification of accumulated circRNA levels by qRT-PCR (C) and circRNA-derived protein expression by western blot (D-E), at indicated timepoints post-transfection.

CircVac, a viral-based expression vector for durable antigen expression

The enhanced stability of circular RNA over mRNA has garnered interest in the development of circular RNA based therapies for disorders where durable protein expression may lead to favourable outcomes, such as vaccination. Viruses are effective vehicles for delivery of transgenes and have been engineered into vaccine vectors capable of stimulating immune responses against the antigens from the encoded transgenes. Here, we describe the generation of our proprietary circVac vector system, a viral vector for delivery of circRNA-encoded antigens (**Fig. 4A**). We demonstrate successful expression of a model antigen from our circVac vector and observe superior expression of the antigen at later timepoints (**Fig. 4B-C**). We hypothesize that the combination of intrinsic immunostimulatory features of the circVac vector and prolonged antigen expression from the circular RNA template, could improve vaccine efficacy.

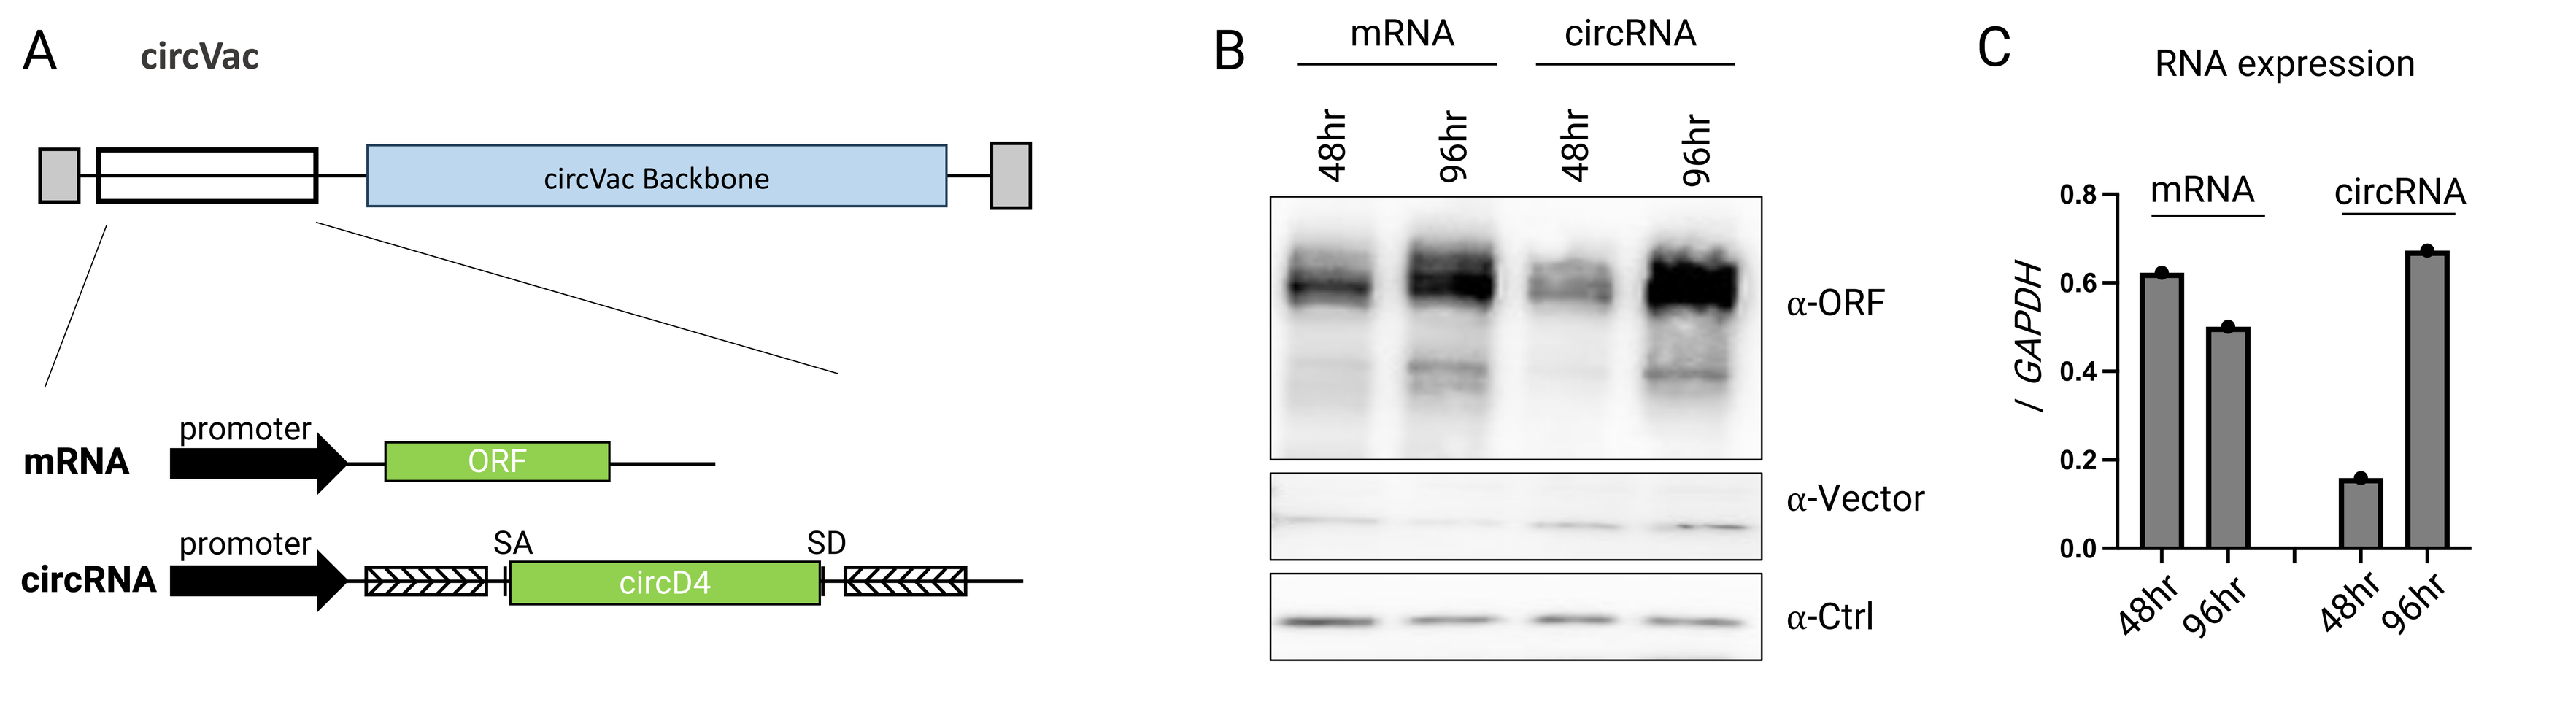
A
circVac
circVac Backbone
mRNA
promoter
ORF
circRNA
promoter SA SD
circD4
B
mRNA circRNA
48hr 96hr 48hr 96hr
α-ORF
α-Vector
α-Ctrl
C
RNA expression
mRNA circRNA
/ GAPDH
48hr 96hr 48hr 96hr

Figure 4: circVac, a viral vector-based system for durable antigen expression. A) Schematic representation of circVac vector backbone (upper). Schematics of mRNA and circRNA expression cassettes, as denoted, inserted within circVac backbone at denoted position (lower). **B)** Antigen protein expression of antigen payload at early (48hr) and late (96hr) timepoints was assessed by western blot. Vector protein expression was used as a proxy to assess viral infection and beta-actin was used as a loading control. **C)** Quantification of antigen RNA expression from circVac and mRNA encoding viral vector, was assessed by RT-qPCR and normalised to *GAPDH* mRNA.

High yield circAde expression is dependent on position of circRNA cassette

In addition to vaccination, viruses are attractive vectors for cancer treatment. Conditionally replicating, also known as oncolytic viruses, have been shown to be an effective form of immunotherapy for treatment of cancer due to their ability to selectively replicate in cancer cells resulting in tumour cell lysis and enhanced immune response. Here, we describe the generation of our proprietary circAde vector, a conditionally-replicating viral vector capable of delivering circular RNA to solid tumours. Effective expression of circular RNA from circAde vectors depends highly on site of integration. From nine initial genome designs, only six circAdes were functional (**Fig. 5A**). To further assess the effect of circRNA integration site on viral fitness the ratio of infectious units per viral particles were assessed (**Fig. 5B-C**). Additionally, dramatic differences in circRNA-based protein expression were observed for the functional circAdes (**Fig. 5D-E**). Comparing circRNA-derived protein expression to IU/VP ratio, higher protein expression was generally associated with lower IU/VP ratios (**Fig. 5F**).

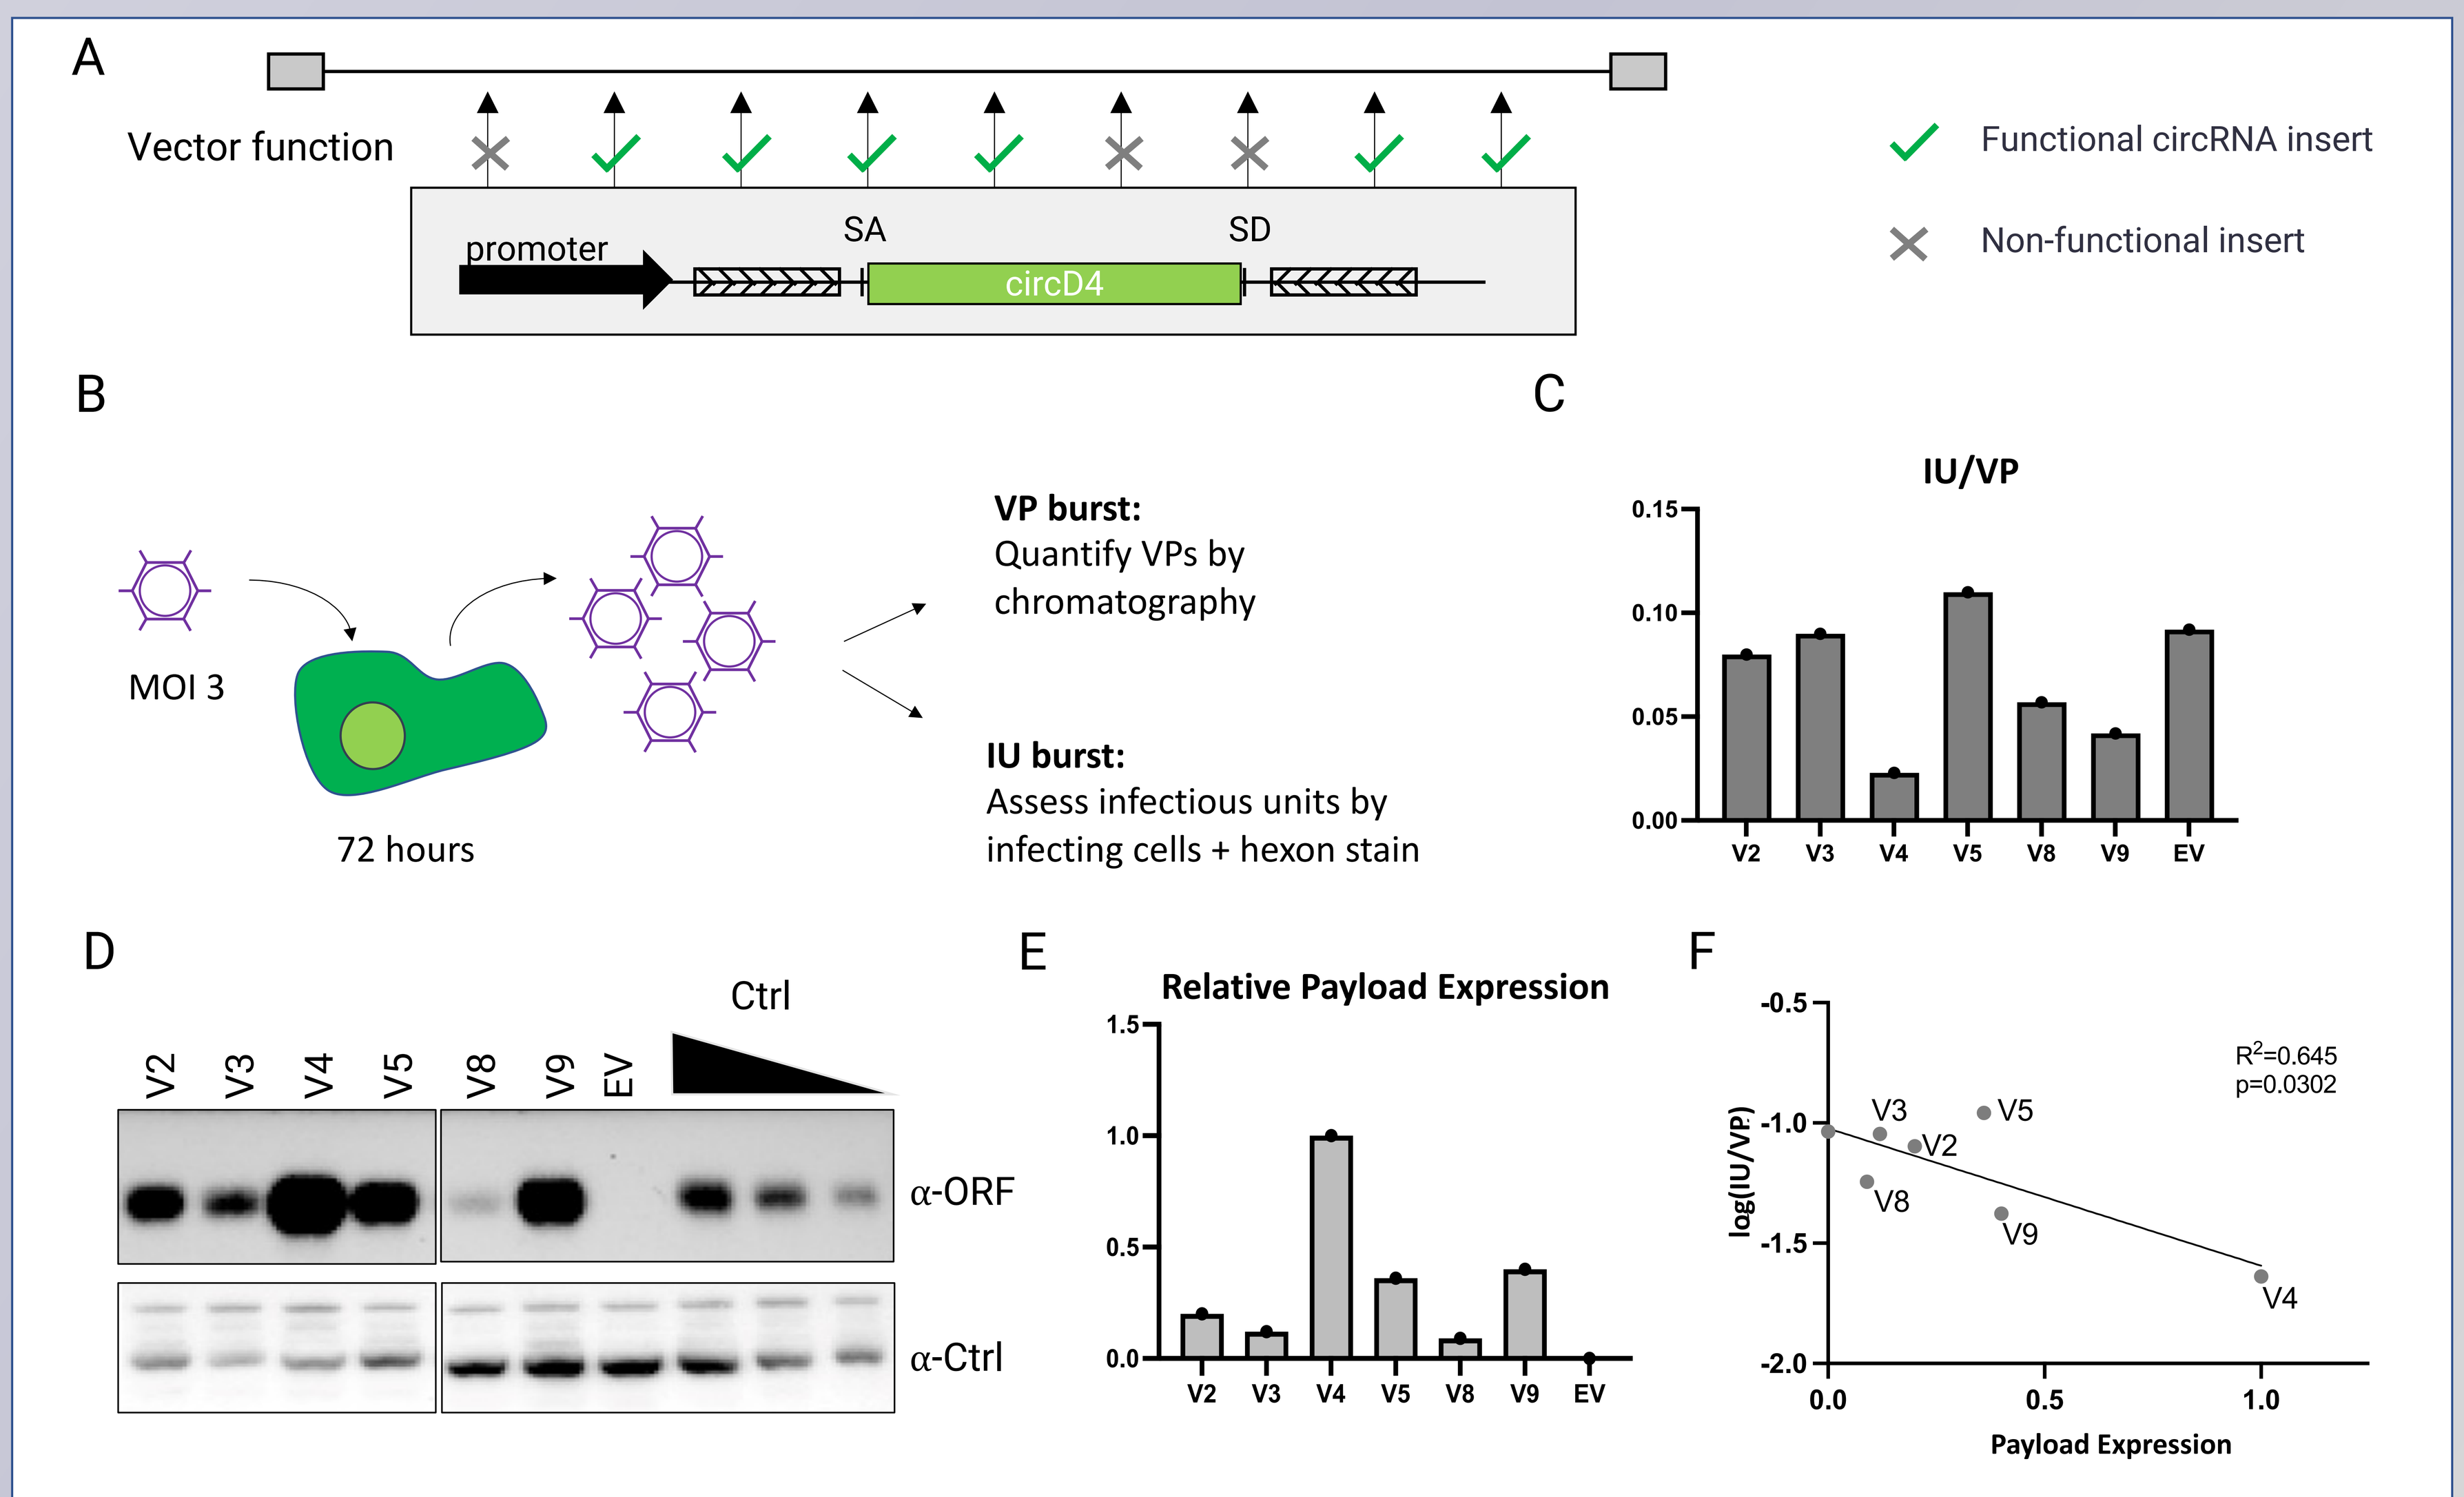
A
Vector function
promoter SA SD
circD4
Functional circRNA insert
Non-functional insert
B
MOI 3
72 hours
VP burst:
Quantify VPs by chromatography
IU burst:
Assess infectious units by infecting cells + hexon stain
C
IU/VP
V2 V3 V4 V5 V8 V9 EV
D
V2 V3 V4 V5 V8 V9 EV
Ctrl
α-ORF
α-Ctrl
E
Relative Payload Expression
V2 V3 V4 V5 V8 V9 EV
F
R^2=0.645
p=0.0302
log(IU/VP)
Payload Expression
V3 V5 V2 V8 V9 V4

Figure 5: Impact on genome integration site strategy on vector functionality and circRNA biogenesis. A) Schematic representation of 9 different integration sites tested. The green checkmark denotes that the circAde vectors were rescued and viable, whereas for the sites with grey cross, no viable circAdes were obtained. **B**) Schematics depicting experimental set-up for viral particle (VP) and infectious unit (IU) assessment. **C**) Graph depicting IU/VP ratios, where IU was assessed by hexon staining and VP was assessed by chromatography. **D**) Western blot on cells transduced with six different circAdes and empty vector (EV) control using payload-specific antibody (upper panel) or loading control (bottom panel). **E**) Graph depicting quantification of payload relative to loading control. **F**) Scatter plot depicting protein expression relative to log(IU/VP) ratio. Linear regression analysis was performed with GraphPad, and a significant negative correlation (p=0.0302) was observed between payload expression and IU/VP ratios.

Conclusion

1) Choice and composition of IR is crucial for high yield circRNA biogenesis.
2) IRES/ORF positioning is essential for generation of protein-coding circRNAs.
3) Superior circRNA stability facilitates accumulation of circRNA and prolonged protein expression.
4) circVac vector shows high yield antigen expression.
5) circAde functionality and expression is highly dependent on circRNA cassette integration site.

The results support further development of the circRNA expression vectors towards therapies where high and prolonged expression of any gene of interest is warranted.

Acknowledgements

This study was funded by Circio Holding ASA Norway. The authors thank everyone in the company for a great atmosphere and working environment. The authors also thank the MUK group at the Karolinska Institutet for their collaboration. We would also like to thank OD260 for production circAde viruses.

Contact information

eoghan.oleary@circio.com

Copies of this poster can be obtained via the QR code
(for personal use and may not be reproduced without author permission)