ONCOS-102 AND PEMETREXED/CISPLATIN IN PATIENTS WITH UNRESECTABLE MALIGNANT PLEURAL MESOTHELIOMA

Santiago Ponce[1], Santiago Viteri[2], Joaquim Bosch-Barrera[3], Lukasz Kuryk[4], Sylvia Vetrhus[5], Magnus Jäderberg[5], Luis Paz-Ares[1]

[1]Hospital 12 Octubre, Madrid, Spain, [2]Hospital Universitario Dexeus. Grupo QuironSalud. Instituto Oncológico Dr Rosell. Barcelona, Spain [3]Institut Català D'Oncologia (I.C.O), Girona, Spain, [4]Targovax OY, Helsinki, Finland, [5]Targovax ASA, Oslo, Norway

BACKGROUND

- Mesothelioma is a rare cancer that is almost always caused by exposure to asbestos. The prognosis for patients with mesothelioma is poor. Median survival ranges between 21 months from diagnosis for patients with early (Stage I) disease, and 12 months for patients with advanced (Stage IV) disease. The American Cancer Society estimates the relative 5-year survival rate for mesothelioma to be between 5% and 10%. Treatment options for patients with mesothelioma are limited. Patients may undergo surgery or radiotherapy but these are not curative. Most patients are in-operable and given chemotherapy, usually as a combination of pemetrexed and cisplatin or carboplatin.

- Virotherapy can assist in breaking the immune tolerance acquired by tumours and activate the immune system against tumour cells[1]. Several oncolytic viruses are in development and one, Imlygic, has been approved in advanced melanoma.

- ONCOS-102 is a serotype 5 adenovirus that is genetically modified to facilitate anti-tumour activity; See Figure 1.

- Synergism between ONCOS-102 and chemotherapy (pemetrexed, cisplatin, carboplatin) has been shown in humanized malignant mesothelioma animal model[2].

Figure 1. The structure of ONCOS-102

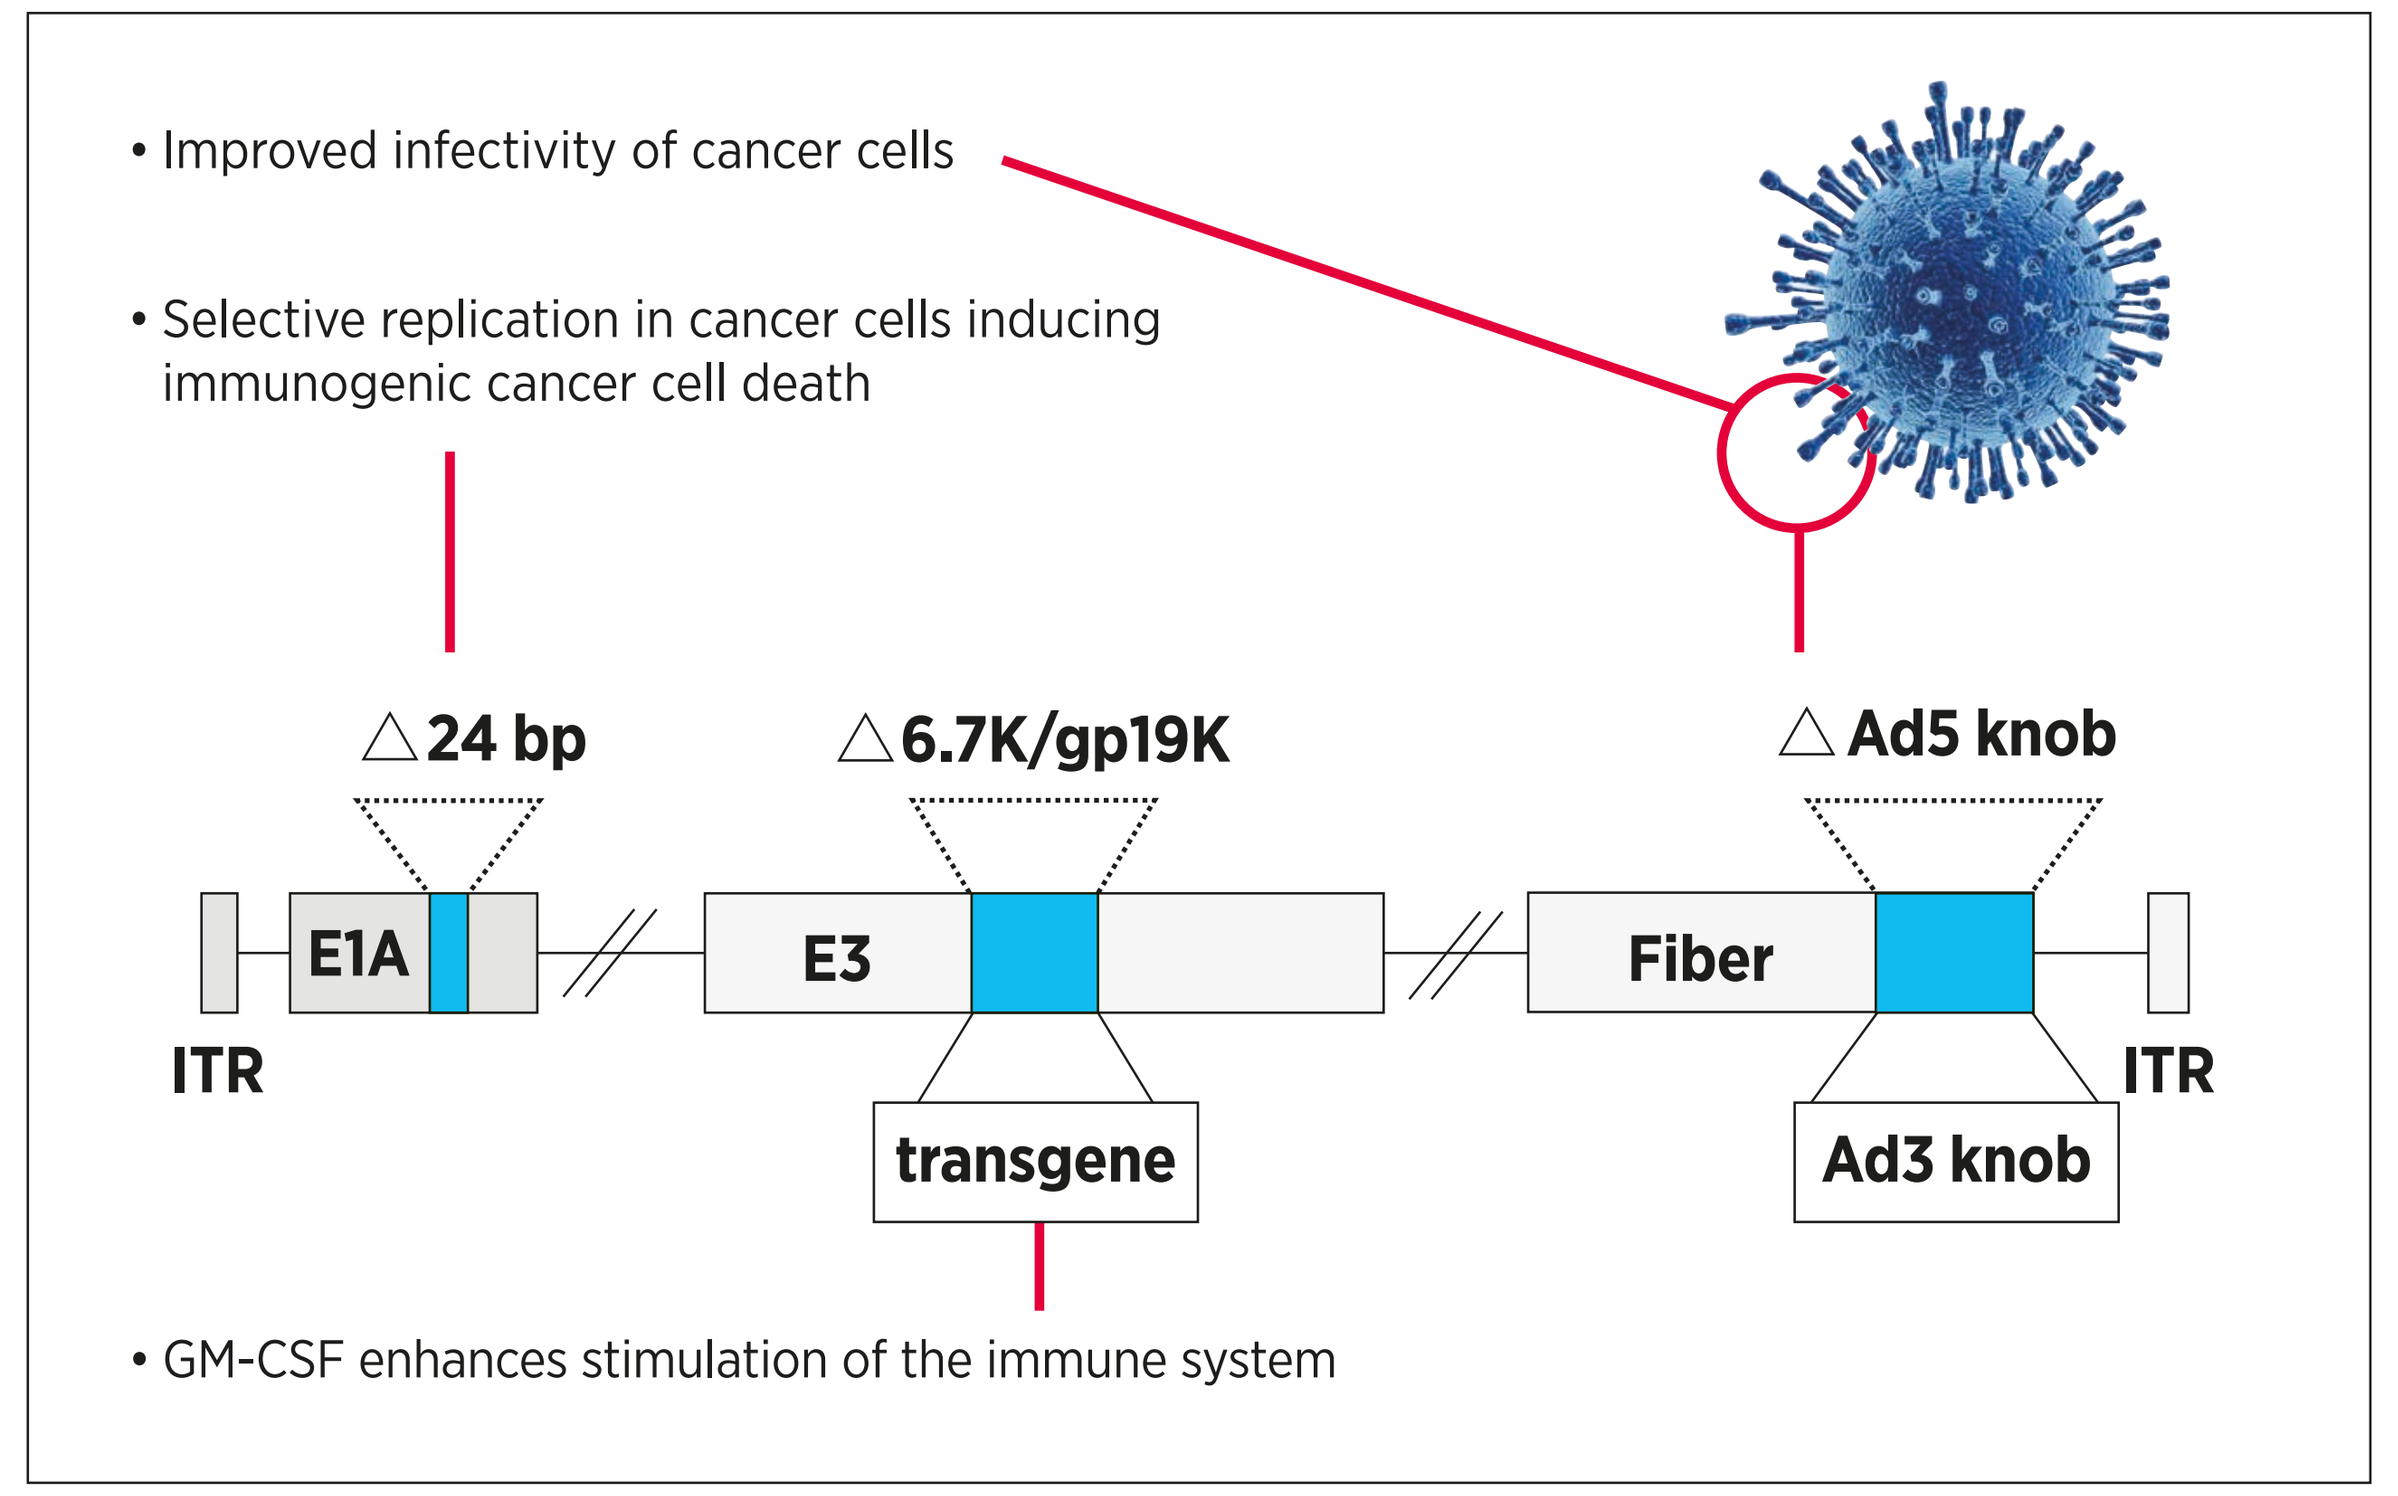
• Improved infectivity of cancer cells
• Selective replication in cancer cells inducing immunogenic cancer cell death
△24 bp
△6.7K/gp19K
△Ad5 knob
E1A
E3
Fiber
ITR
ITR
transgene
Ad3 knob
• GM-CSF enhances stimulation of the immune system

ONCOS-102 mechanism of action is shown in figure 2.

Figure 2. ONCOS-102 mechanism of action

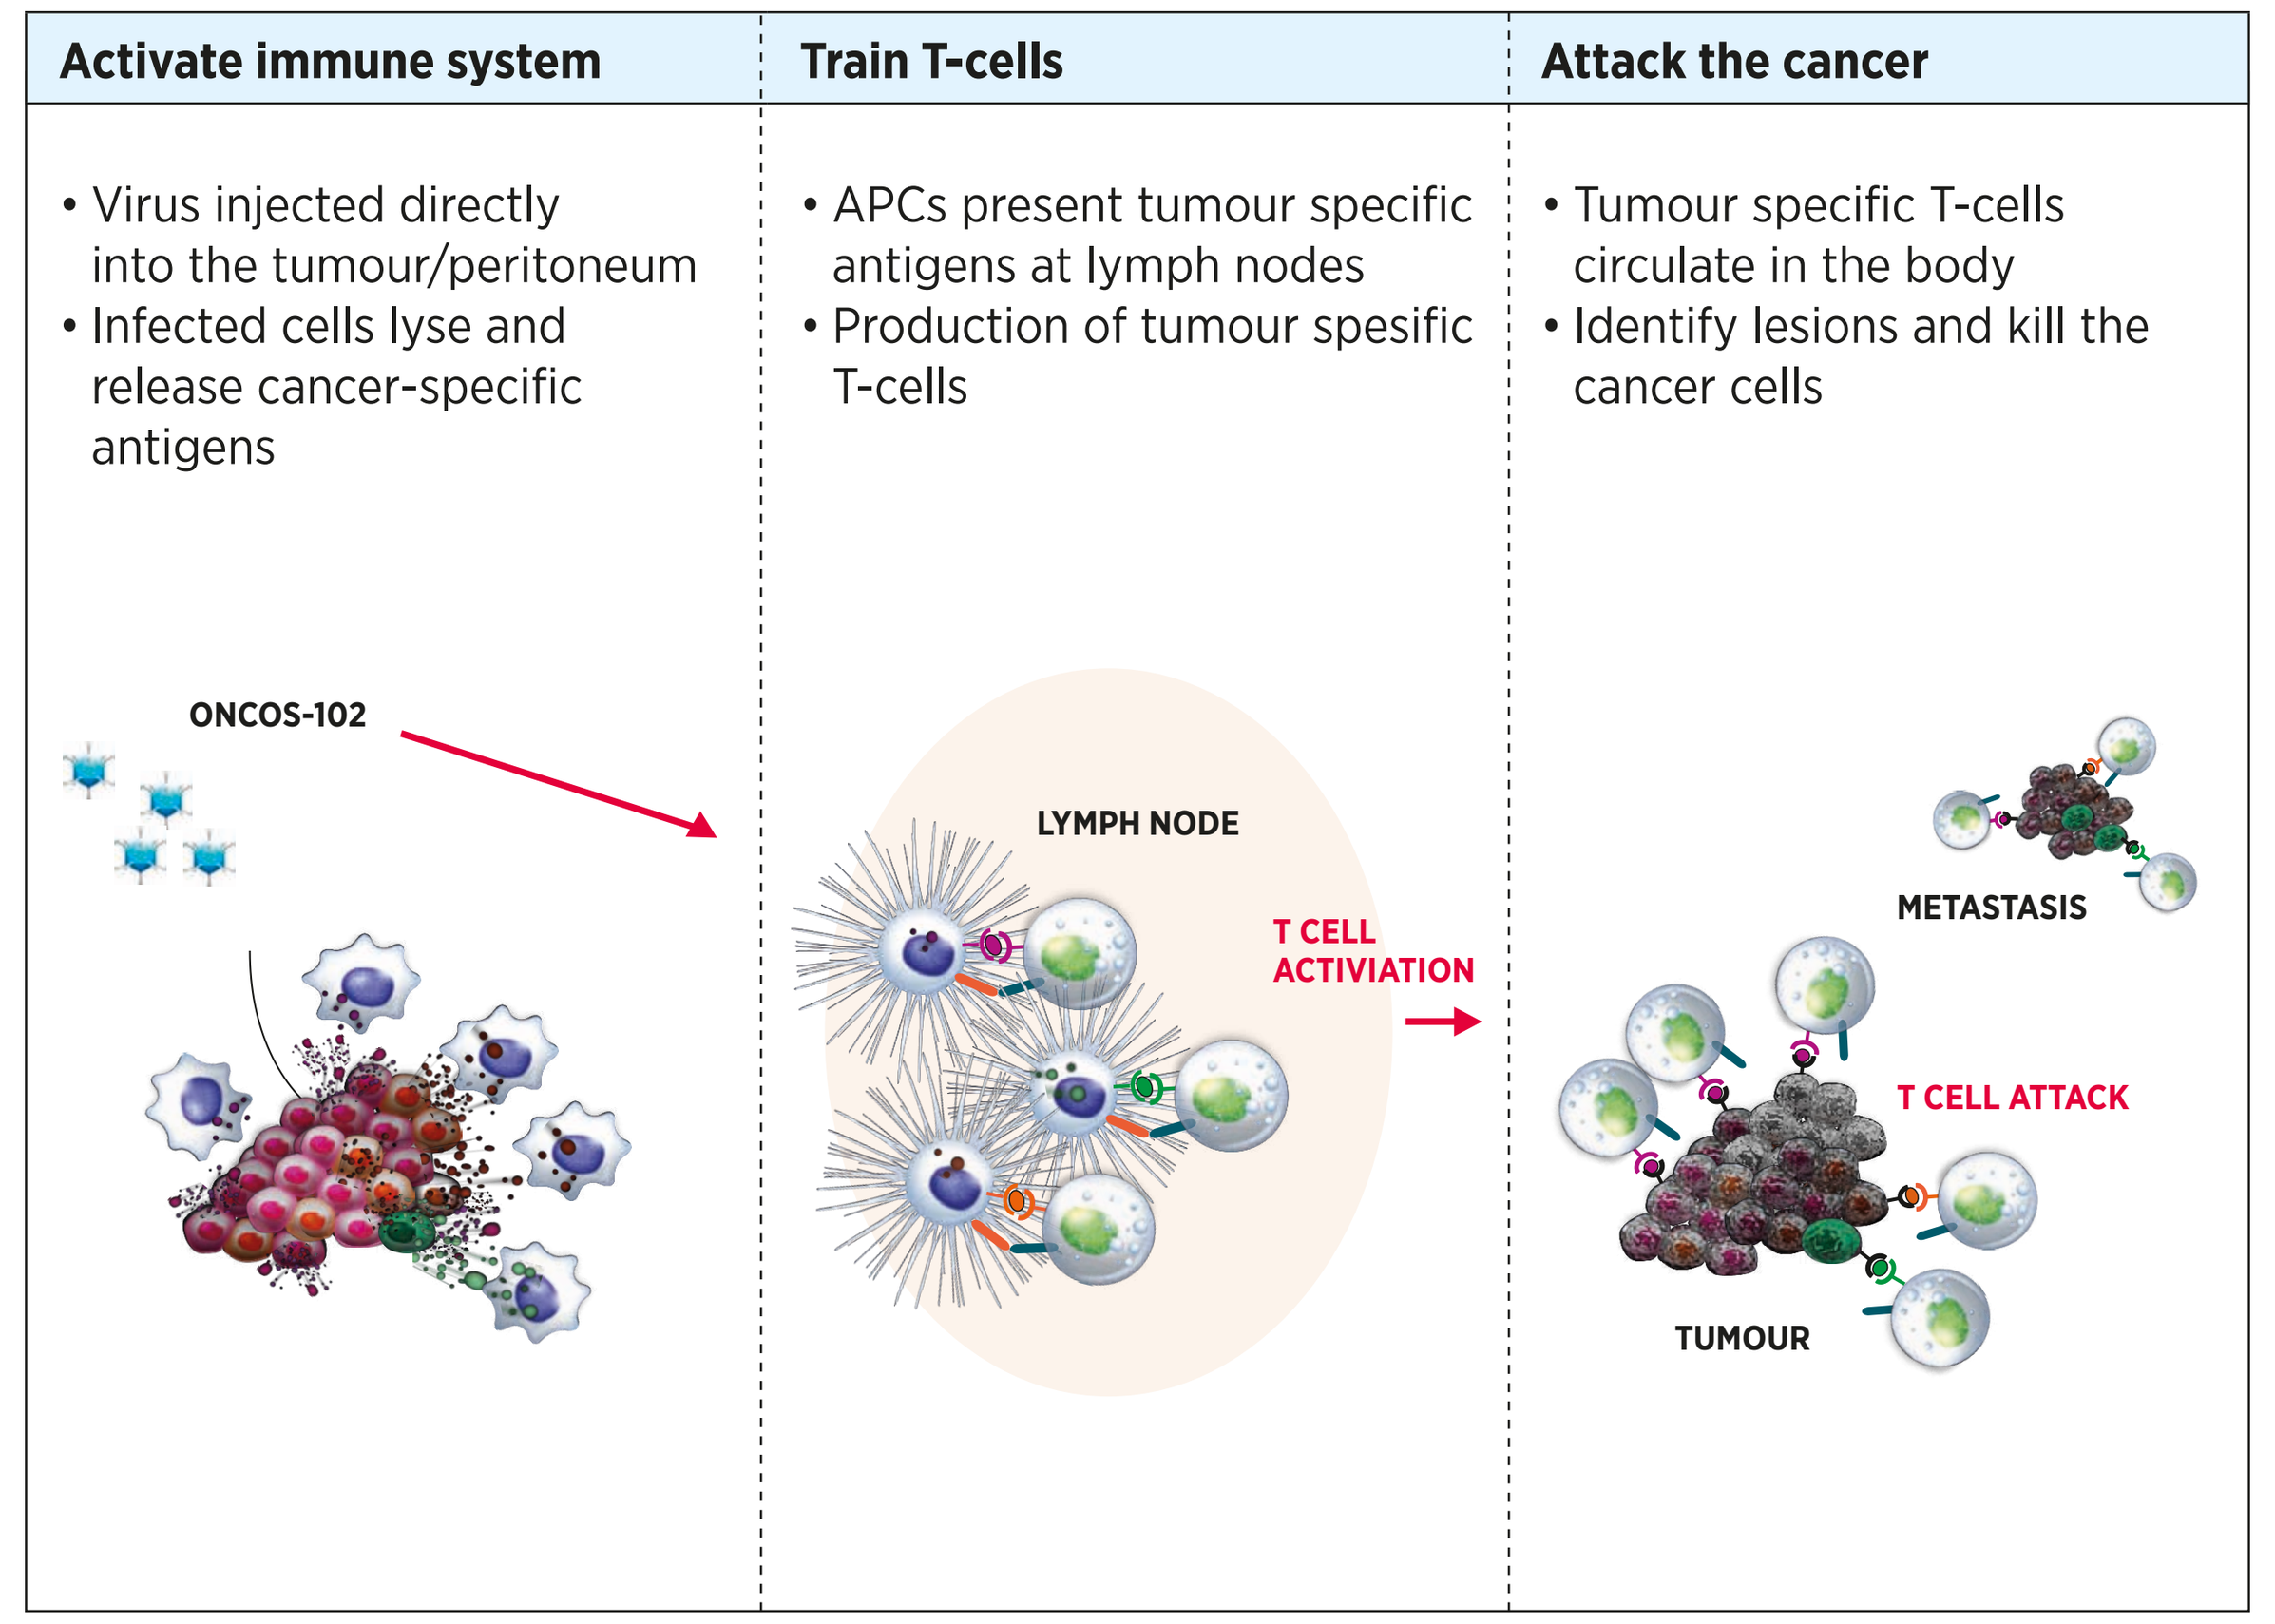
Activate immune system
Train T-cells
Attack the cancer
• Virus injected directly into the tumour/peritoneum
• Infected cells lyse and release cancer-specific antigens
• APCs present tumour specific antigens at lymph nodes
• Production of tumour spesific T-cells
• Tumour specific T-cells circulate in the body
• Identify lesions and kill the cancer cells
ONCOS-102
LYMPH NODE
T CELL ACTIVIATION
METASTASIS
T CELL ATTACK
TUMOUR

Clinical experience

A Phase I clinical trial (NCT01598129) of ONCOS-102 with cyclophosphamide (CPO) has been completed. Twelve patients with various refractory injectable solid tumours were enrolled with assessment of safety, disease progression and immune activation[3].

SAFETY Patients were treated with ONCOS-102 at doses of up to 3 x 10^{11} VP for a duration of 6 months. Most of the AEs reported in the Phase I study were Grade 1 or 2. Grade 3 AEs were reported in 6 patients and there were no Grade 4 AEs.

DISEASE PROGRESSION 40% (4/10 evaluable patients) developed stabile disease (SD) at 3 months. Two of the patients had mesothelioma:

Patient A: 63-year-old male with asbestosis-related pleural disease entered the study after not responding to chemotherapy. He was discontinued at 3 months due to progression.

Patient B: 68-year-old male also with asbestosis-related pleural disease, entered the study after progression despite two different chemotherapy treatments. He completed the 6 month study and was called back for a routine assessment 6 weeks later when PET imaging showed a 47% reduction of metabolic activity of his tumour compared with the 6 month study visit. The patient had not received additional treatments after the study.

IMMUNE ACTIVATION Immune cell analysis was done in blood (PBMC) and tumour (biopsy) at baseline and during treatment. Both patients showed an increase of CD8+ T-cells in response to ONCOS-102 treatment and one of the patients, where IFN-gamma ELISPOT analysis of PBMC was possible, showed an increase of MAGE-A3 CD8+ T-cells thus evidence of tumour specific immune activation.

TRIAL DESIGN

Open-label, parallel group, multicenter study conducted in 2 phases: a randomized Phase II study (n=24) with a non-randomized Phase Ib safety lead-in cohort (n=6).

Treatment schedule for both active arm and control arm are shown in figure 3.

Single cyclophosphamide (CPO) dose followed by intratumoral injection of ONCOS-102 at 3×10^{11} viral particles (VPs). Pemetrexed (500mg/m2)/ cisplatin(75mg/m^2) is given every 3 weeks for a maximum of 6 cycles.

Figure 3a. Study schedule active arm (ONCOS-102 + pemetrexed/cisplatin)

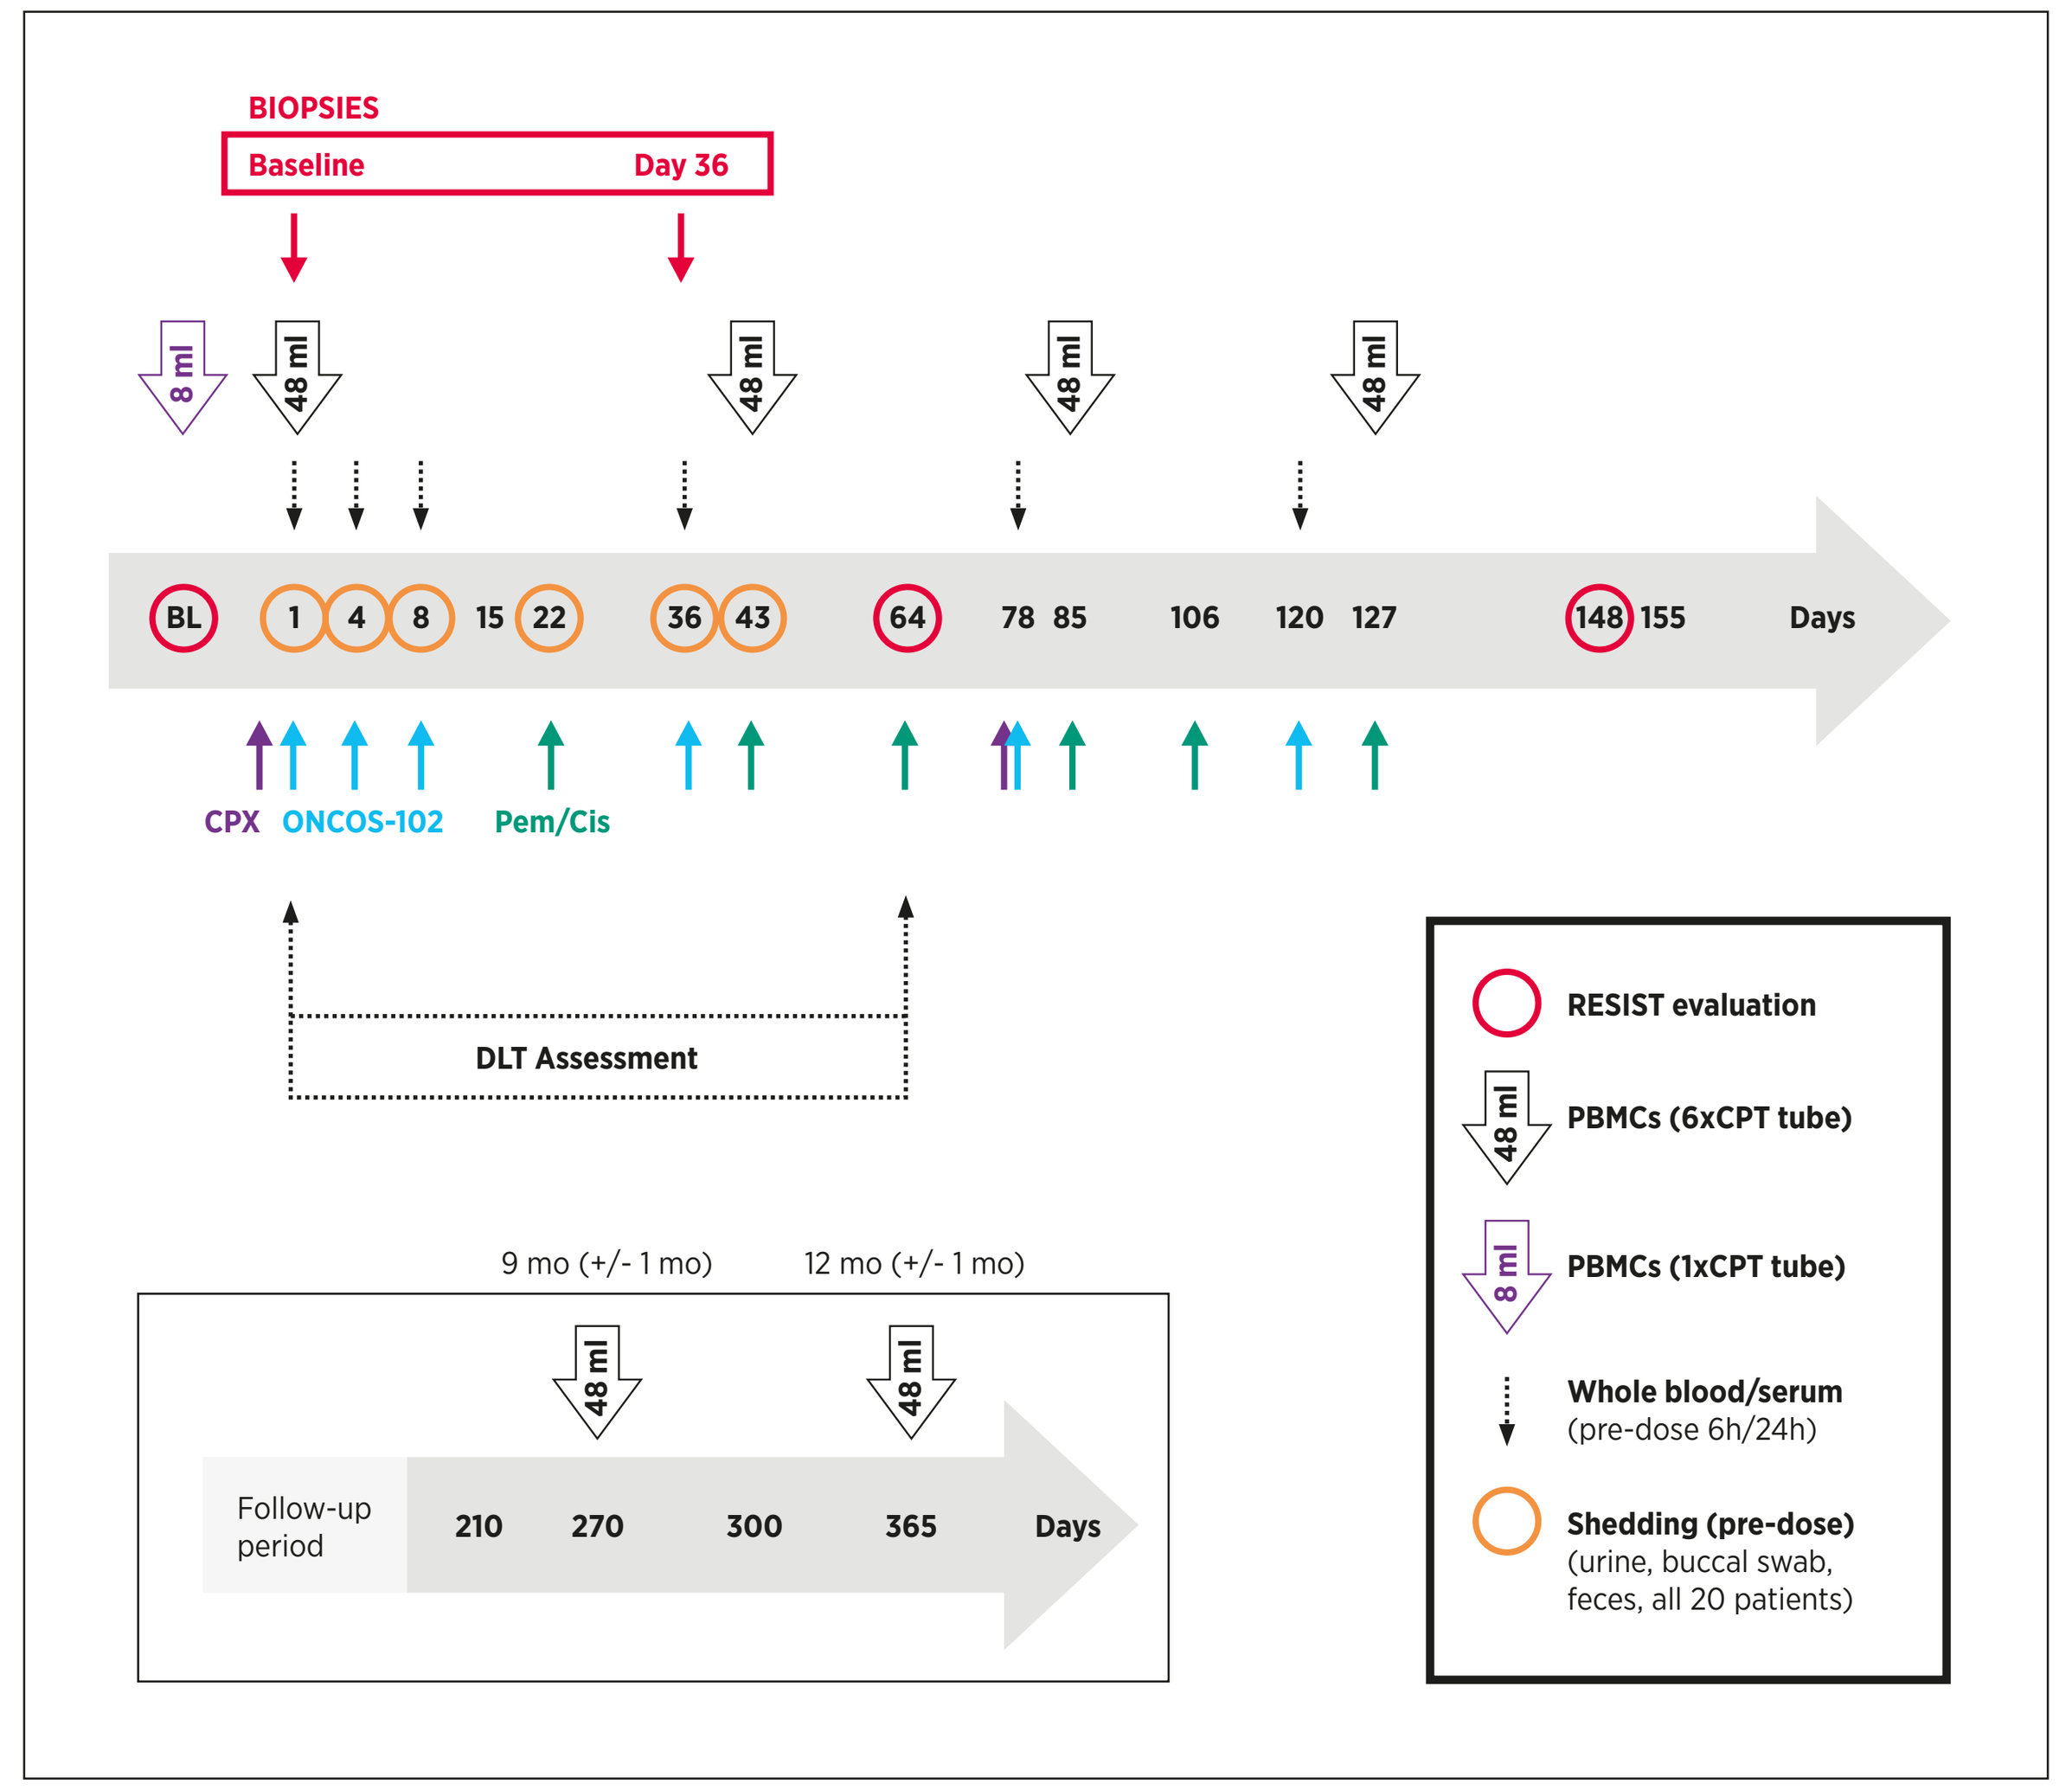
BIOPSIES
Baseline
Day 36
8 ml
48 ml
BL 1 4 8 15 22 36 43 64 78 85 106 120 127 148 155 Days
CPX
ONCOS-102
Pem/Cis
DLT Assessment
9 mo (+/- 1 mo)
12 mo (+/- 1 mo)
Follow-up period
210 270 300 365 Days
RESIST evaluation
PBMCs (6xCPT tube)
PBMCs (1xCPT tube)
Whole blood/serum (pre-dose 6h/24h)
Shedding (pre-dose) (urine, buccal swab, feces, all 20 patients)

Figure 3b. Study schedule control arm (pemetrexed/cisplatin)

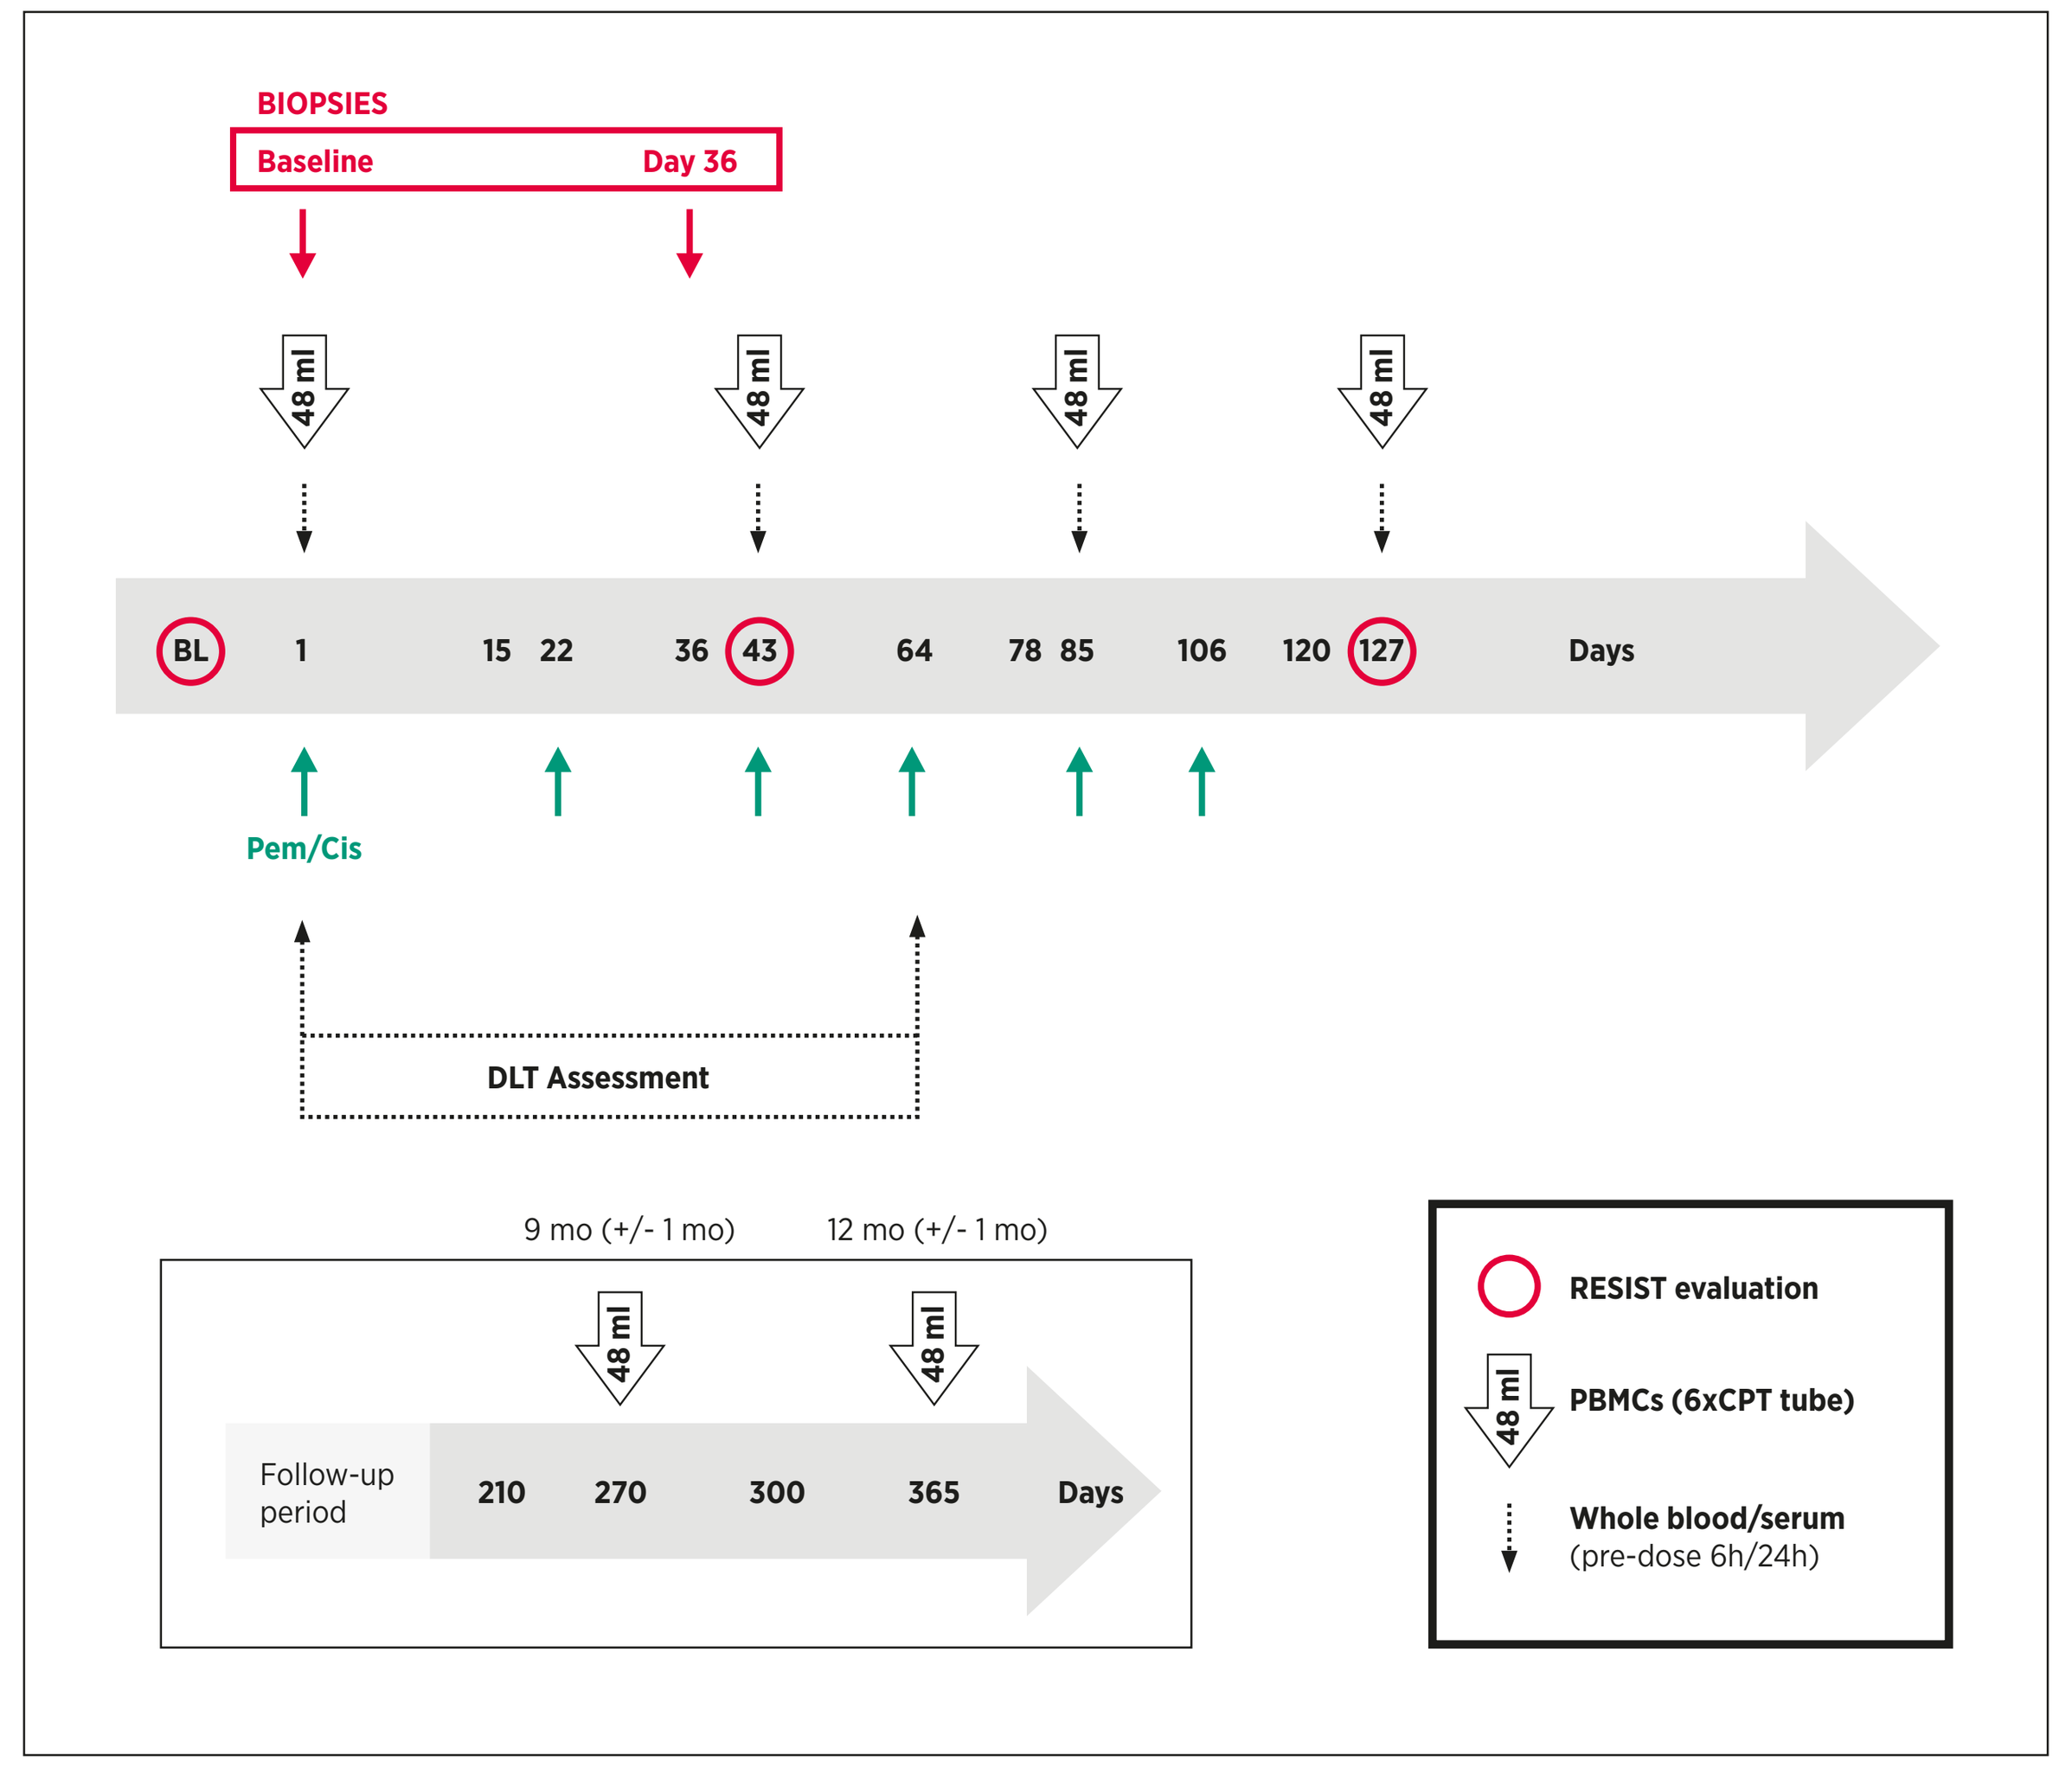
BIOPSIES
Baseline
Day 36
48 ml
BL 1 15 22 36 43 64 78 85 106 120 127 Days
Pem/Cis
DLT Assessment
9 mo (+/- 1 mo)
12 mo (+/- 1 mo)
Follow-up period
210 270 300 365 Days
RESIST evaluation
PBMCs (6xCPT tube)
Whole blood/serum (pre-dose 6h/24h)

Objective

To compare safety, immune response and efficacy of ONCOS-102 + pemetrexed/ cisplatin vs pemeterxed/cisplatin

Main criteria for inclusion

- Histologically confirmed unresectable (advanced) malignant pleural mesothelioma in patients who are not candidates for curative surgery and for whom therapy with pemetrexed/cisplatin is considered appropriate.
 - > This includes patients who are naïve to chemotherapy,
 - > and those who have already received pemetrexed/cisplatin to which their tumour initially responded, but they have relapsed after at least 6 months.
- Measurable disease according to Response Evaluation in Solid Tumour (RECIST) 1.1.
- Tumour must be accessible to intratumoural (i.t.) injections and to tumour core needle biopsy or thoracoscopy for tissue sampling and immunohistochemistry analysis.
- Acceptable liver, renal, and haematological functions.

Main criteria for exclusion

- Receipt of oncolytic virus treatment, or vaccination with a vaccine containing live virus within 4 weeks before Day 1.
- Use of significant immunosuppressive medication, including high dose corticosteroid (defined as the equivalent of >10 mg/day prednisone) within 4 weeks before Day 1.
- Known brain metastases.
- History of malignant tumour, unless the patient has been without evidence of disease for at least 3 years, or the tumour was a non-melanoma skin tumour, cervical carcinoma in situ, or prostatic carcinoma in situ.

Biological sample collection

A number of biological samples will be collected at different timepoints during the course of the study (see Figure 3a and 3b).

- Core biopsies - to understand the mechanism of action of the therapy and to generate hypotheses on predictive and prognostic factors
- Blood samples - for analysis of biological and immunological correlates, e.g., virus replication, cellular responses and humoral responses
- Shedding samples: Saliva, Urine and faeces

Follow-up

The study is completed after 6 months, thereafter patients will be followed every 3 months until end of life for survival and further cancer therapy.

Study Status

Three active sites in Spain, additional sites being added. Submission process ongoing in France and Italy. Approximately 10 sites in total needed.

Enrolment in safety phase is ongoing.

No safety concern reported by Data Safety Monitoring Board following review of safety data from the first three patients.

REFERENCES

1 Qiao et al,; Nat Med; 1; 37-44 (2008)
2 Kuryk et al.; Int. J. Cancer; 139, 1883–1893 (2016)
3 Ranki et al.; Journal for ImmunoTherapy of Cancer; 4:17; (2016)

ClinicalTrials.gov. Identifier: NCT02879669

Design: fetetyper.no

www.targovax.com